REGRESSION OF FIBROSIS AND REVERSAL OF CIRRHOSIS IN THIOACETAMIDE-INDUCED LIVER FIBROSIS FOLLOWING TREATMENT WITH GALECTIN INHIBITORS

Peter G. Traber*, Hsin Chou**, Eliezer Zomer*, Feng Hong**, Anatole Klyosov*, Maria-Isabel Fiel***, Scott L. Friedman**
* Galectin Therapeutics Inc, Norcross, GA, USA
** Division of Liver Diseases, *** Department of Pathology, Icahn School of Medicine at Mount Sinai, NYC, NY, USA

Introduction:
- Liver fibrosis and cirrhosis are important causes of morbidity and mortality and there are currently no approved medical therapies.
- While the pathogenesis of collagen deposition in the liver is complex, galectin-3 protein has been shown in knockout mouse experiments to be a critical protein in the fibrogenic process.
- Complex carbohydrate drugs with terminal galactose residues derived from plant sources bind to galectin-3 and have therapeutic potential.

Objective:
- To evaluate the efficacy of novel complex carbohydrate drugs that inhibit galectin proteins in the treatment of experimental liver fibrosis and cirrhosis.

Methods:
- Liver fibrosis was induced in rats with intra-peritoneal injections of thioacetamide (TAA).
- Rats were treated with either vehicle as a control or various concentrations of GM-CT-01 (galactomannan) or GR-MD-01/02 (galactoarabino-rhamnogalacturonan (GARG)), which bind galectin-3.

Comparison of effects of two different preparations of GARG demonstrated that GR-MD-02 was more effective than GR-MD-01 on reduction of collagen in TAA-treated rats

Weeks	1	2	3	4	5	6	7	8	9	10	11	12
TAA Rx	150 mg/kg ip twice weekly											
Rx Group												
#1 (n=7) Vehicle Control (ip twice weekly)									1	2	3	4
#2 (n=7) GR-MD-01 (60 mg/kg ip twice weekly)									1	2	3	4
#3 (n=8) GR-MD-01 (60 mg/kg ip twice weekly) + NAC (120 mg/kg 2X/wk)									1	2	3	4
#4 (n=7) GR-MD-02 (60 mg/kg ip twice weekly)									1	2	3	4
#5 (n=7) GR-MD-02 (60 mg/kg twice weekly) + NAC (120 mg/kg 2X/wk)									1	2	3	4

Representative liver sections from each group stained with Sirius red

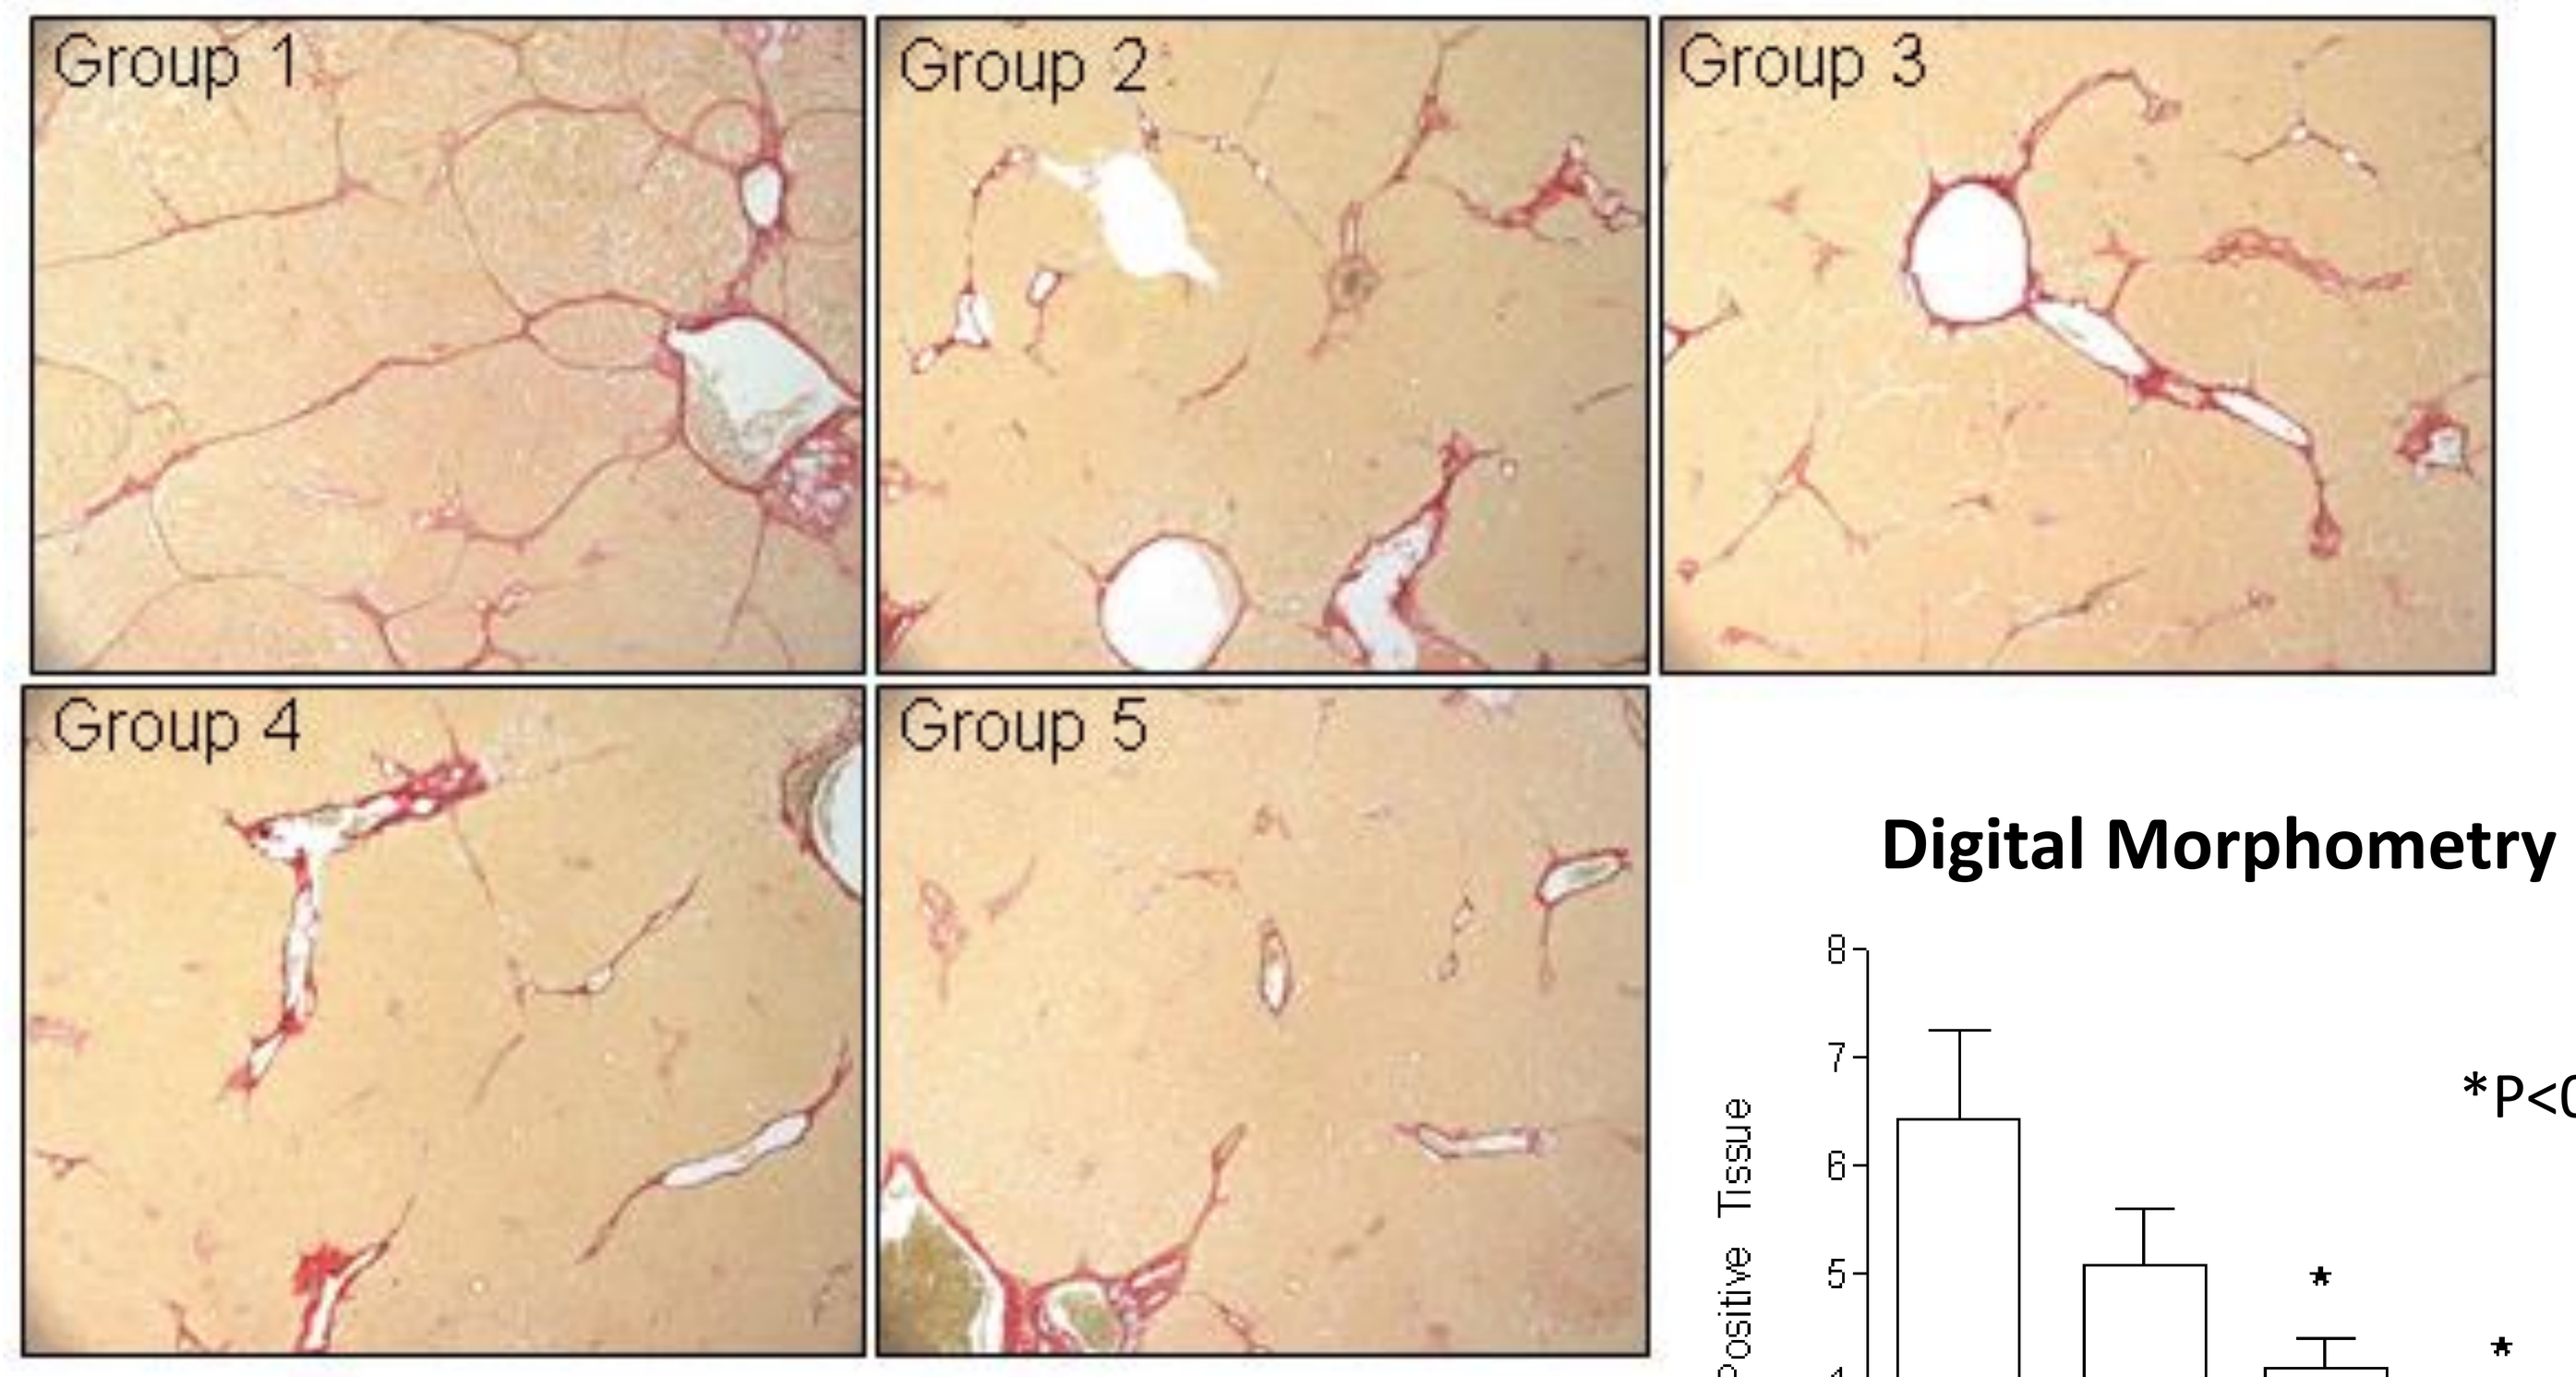
Group 1
Group 2
Group 3
Group 4
Group 5

Digital Morphometry

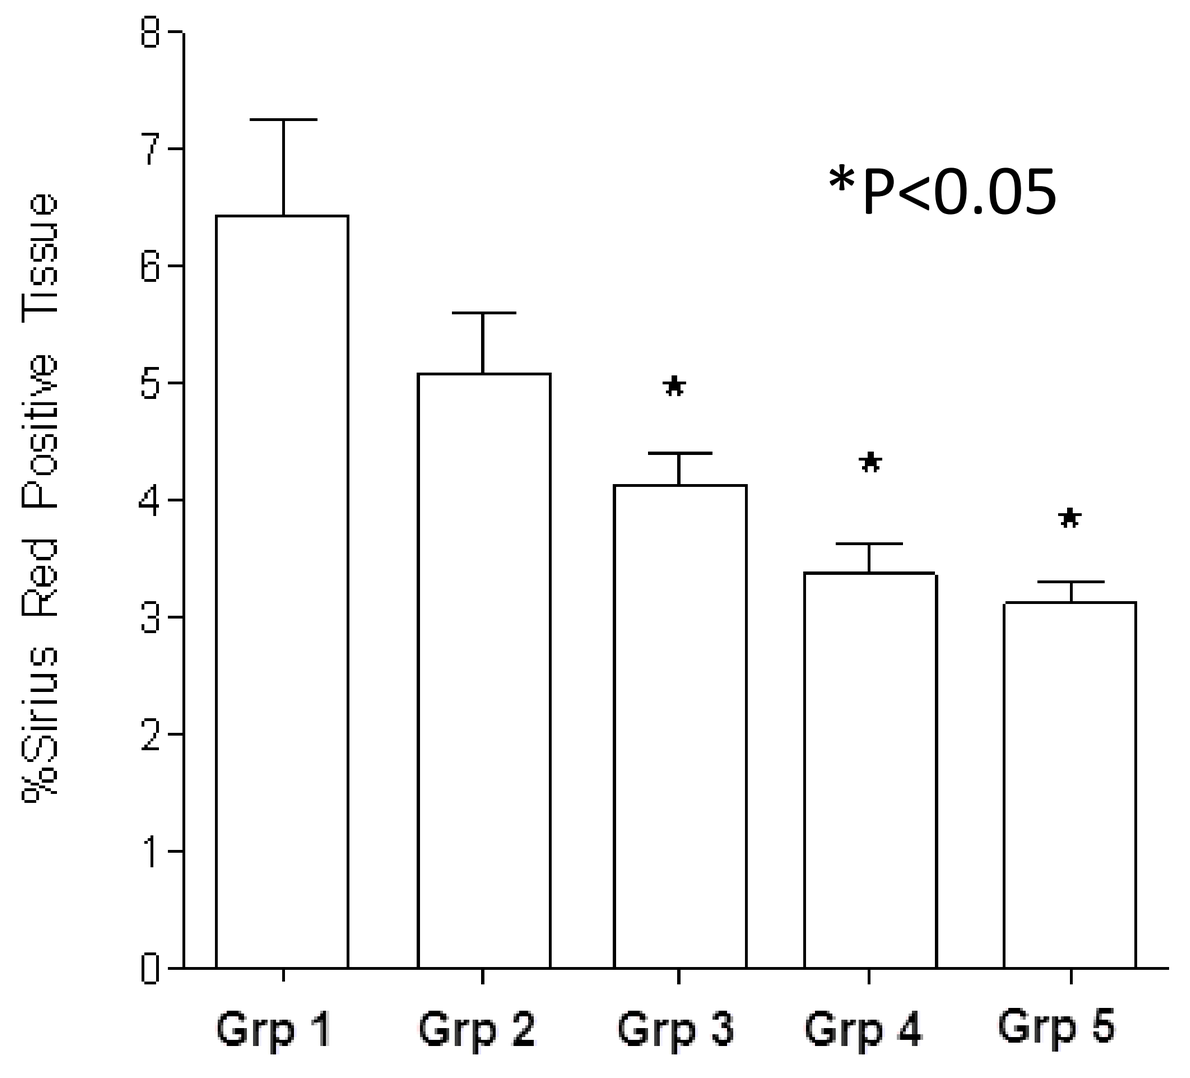
*P<0.05
%Sirius Red Positive Tissue
Grp 1
Grp 2
Grp 3
Grp 4
Grp 5

Note: The addition of N-acetyl cysteine may have enhanced effect of GR-MD-01 but had no the efficacy of GR-MD-02 in reducing fibrosis

Administration of both GR-MD-01 and GM-CT-01 reduced collagen and reversed cirrhosis in rats continuously treated with TAA

Weeks	1	2	3	4	5	6	7	8	9	10	11
TAA Rx	150 mg/kg ip three times weekly										
Rx Group											
#1 (n=10) Vehicle Control (iv 2 x Wk)								1	2	3	4
#2 (n=10) GR-MD-02 (60 mg/kg ip 2 x Wk)								1	2	3	4
#3 (n=10) GR-MD-02 (60 mg/kg ip 1 x Wk)								1	2	3	4
#4 (n=10) GR-MD-02 (90 mg/kg ip 1 x Wk)								1	2	3	4
#5 (n=10) GM-CT-01 (105 mg/kg ip 2 x Wk)								1	2	3	4
#6 (n=10) GM-CT-01 (105 mg/kg ip 1 x Wk)								1	2	3	4
#7 (n=10) GM-CT-01 (180 mg/kg ip 1 x Wk)								1	2	3	4
#8 (n=10) Normal rats without TAA or Drug Treatment											

Representative liver sections from each group stained with Sirius red

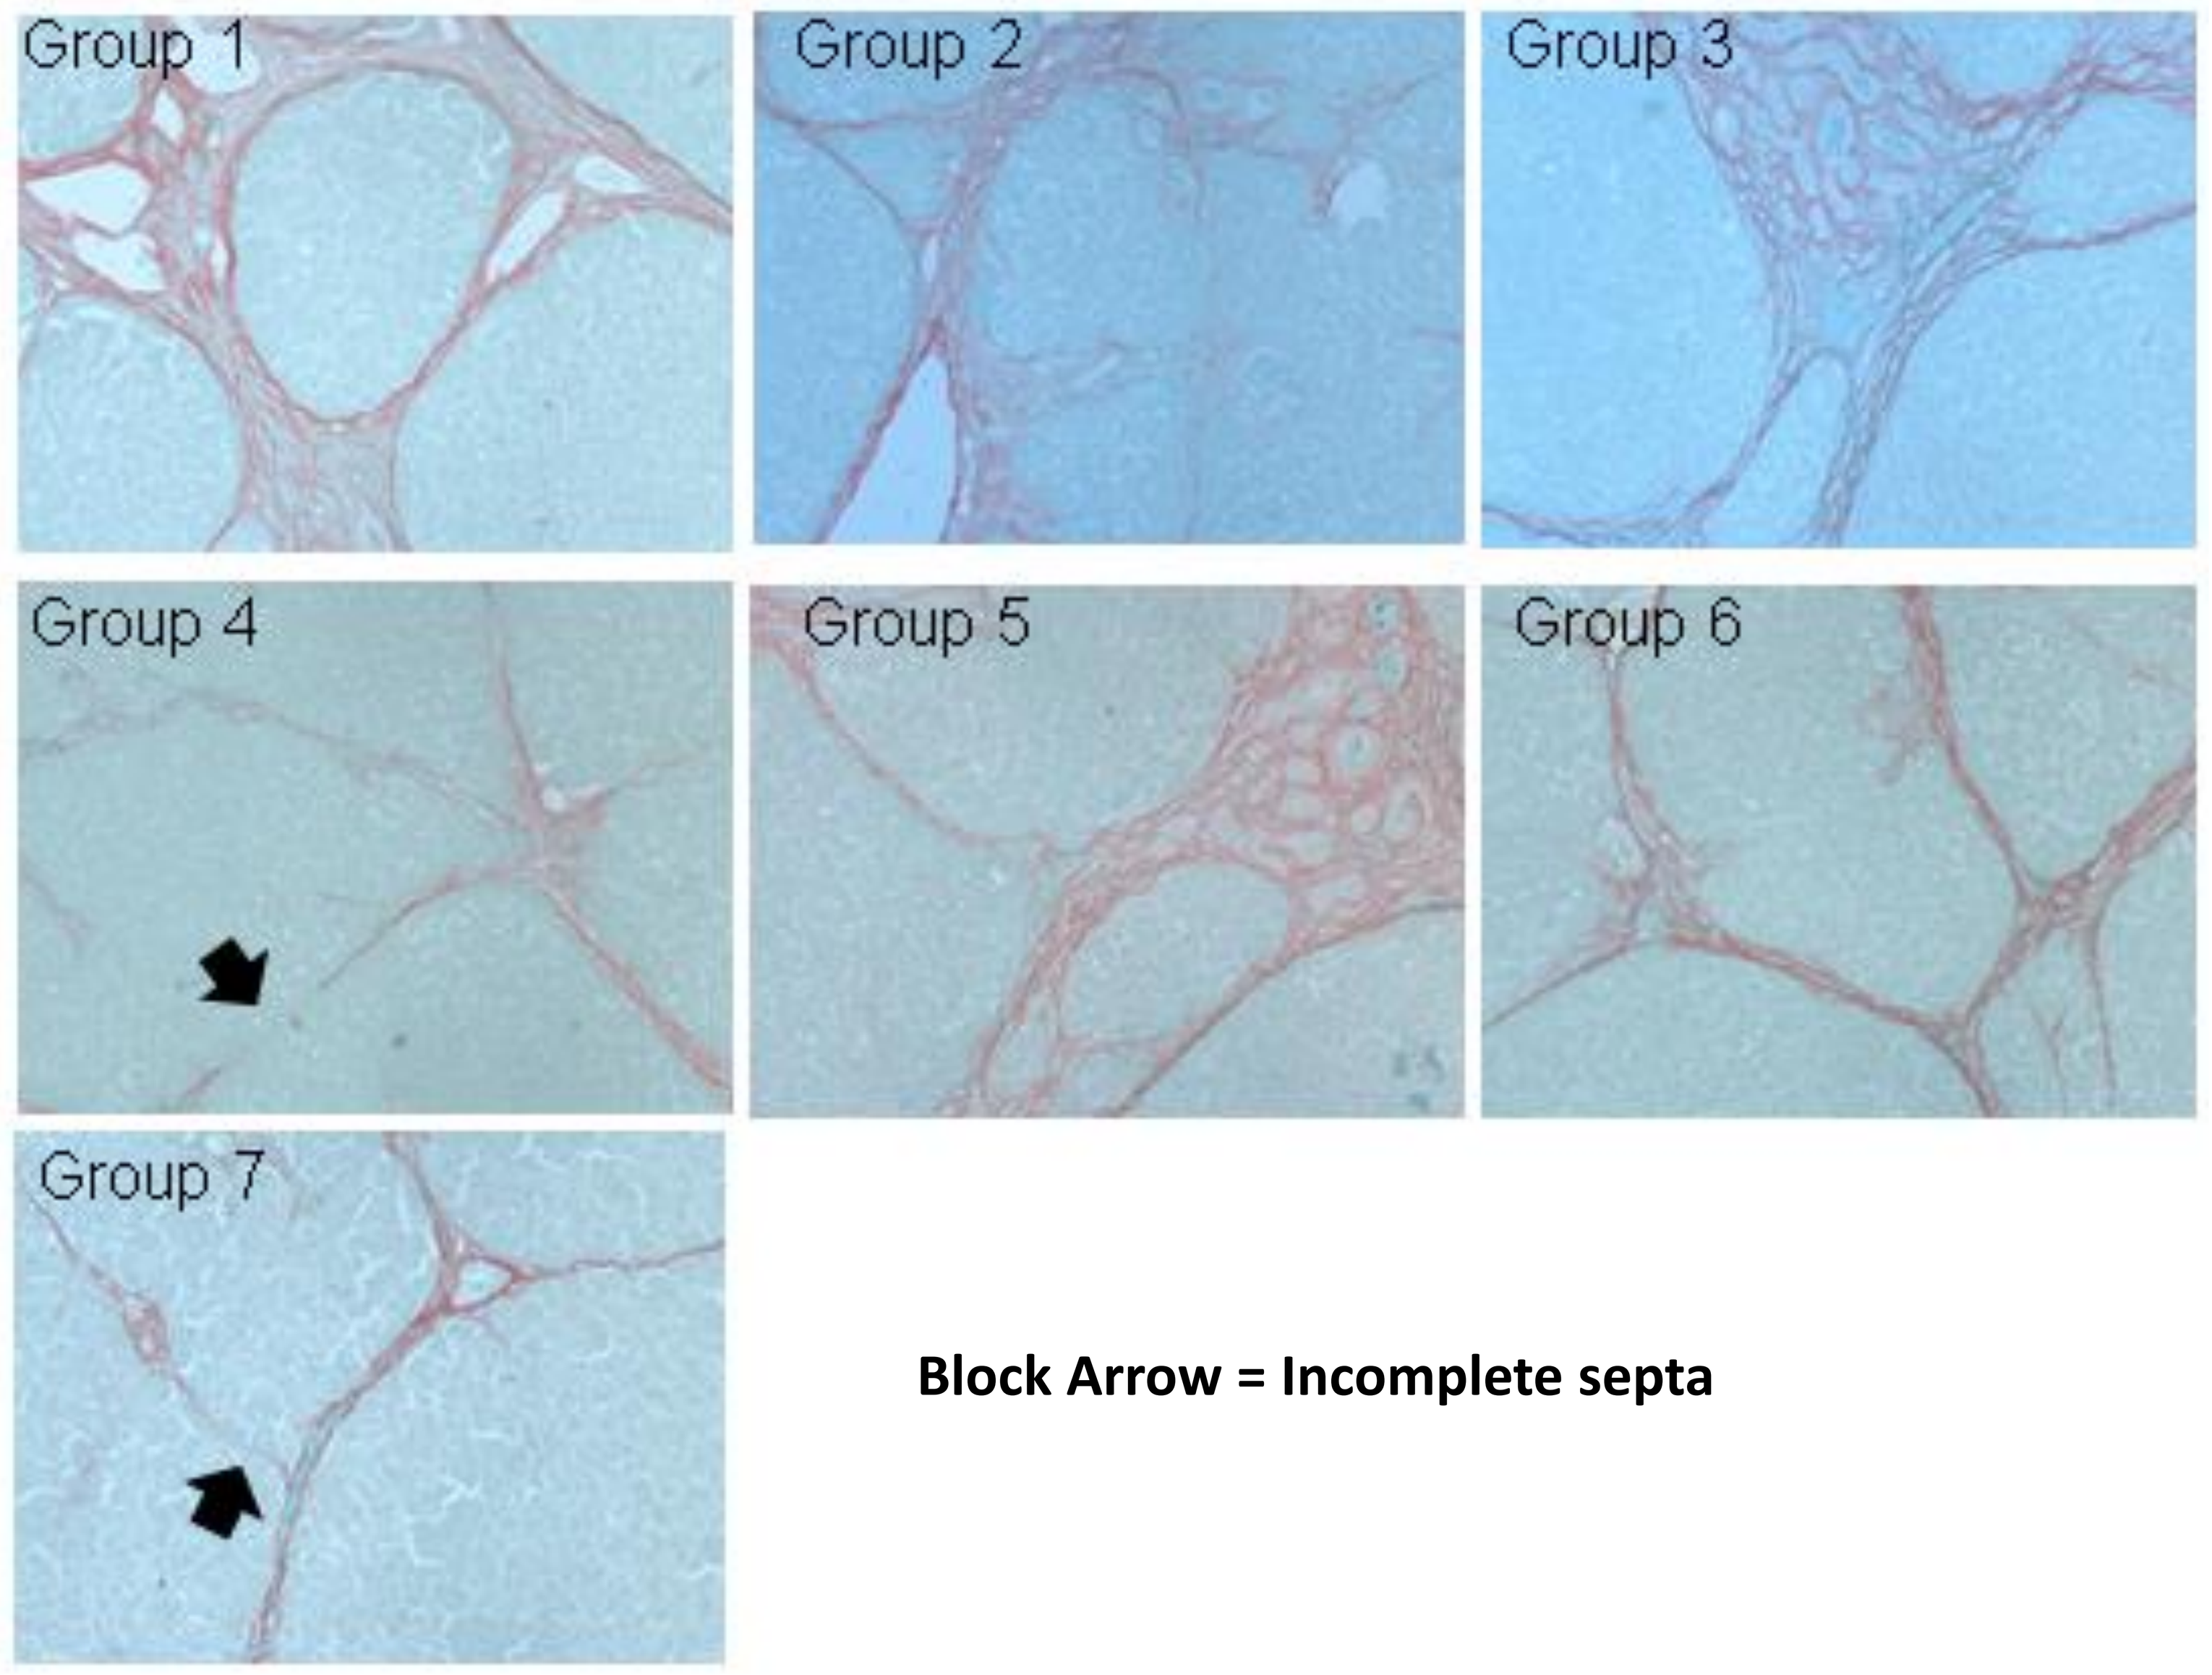
Group 1
Group 2
Group 3
Group 4
Group 5
Group 6
Group 7

Block Arrow = Incomplete septa

Digital Morphometry

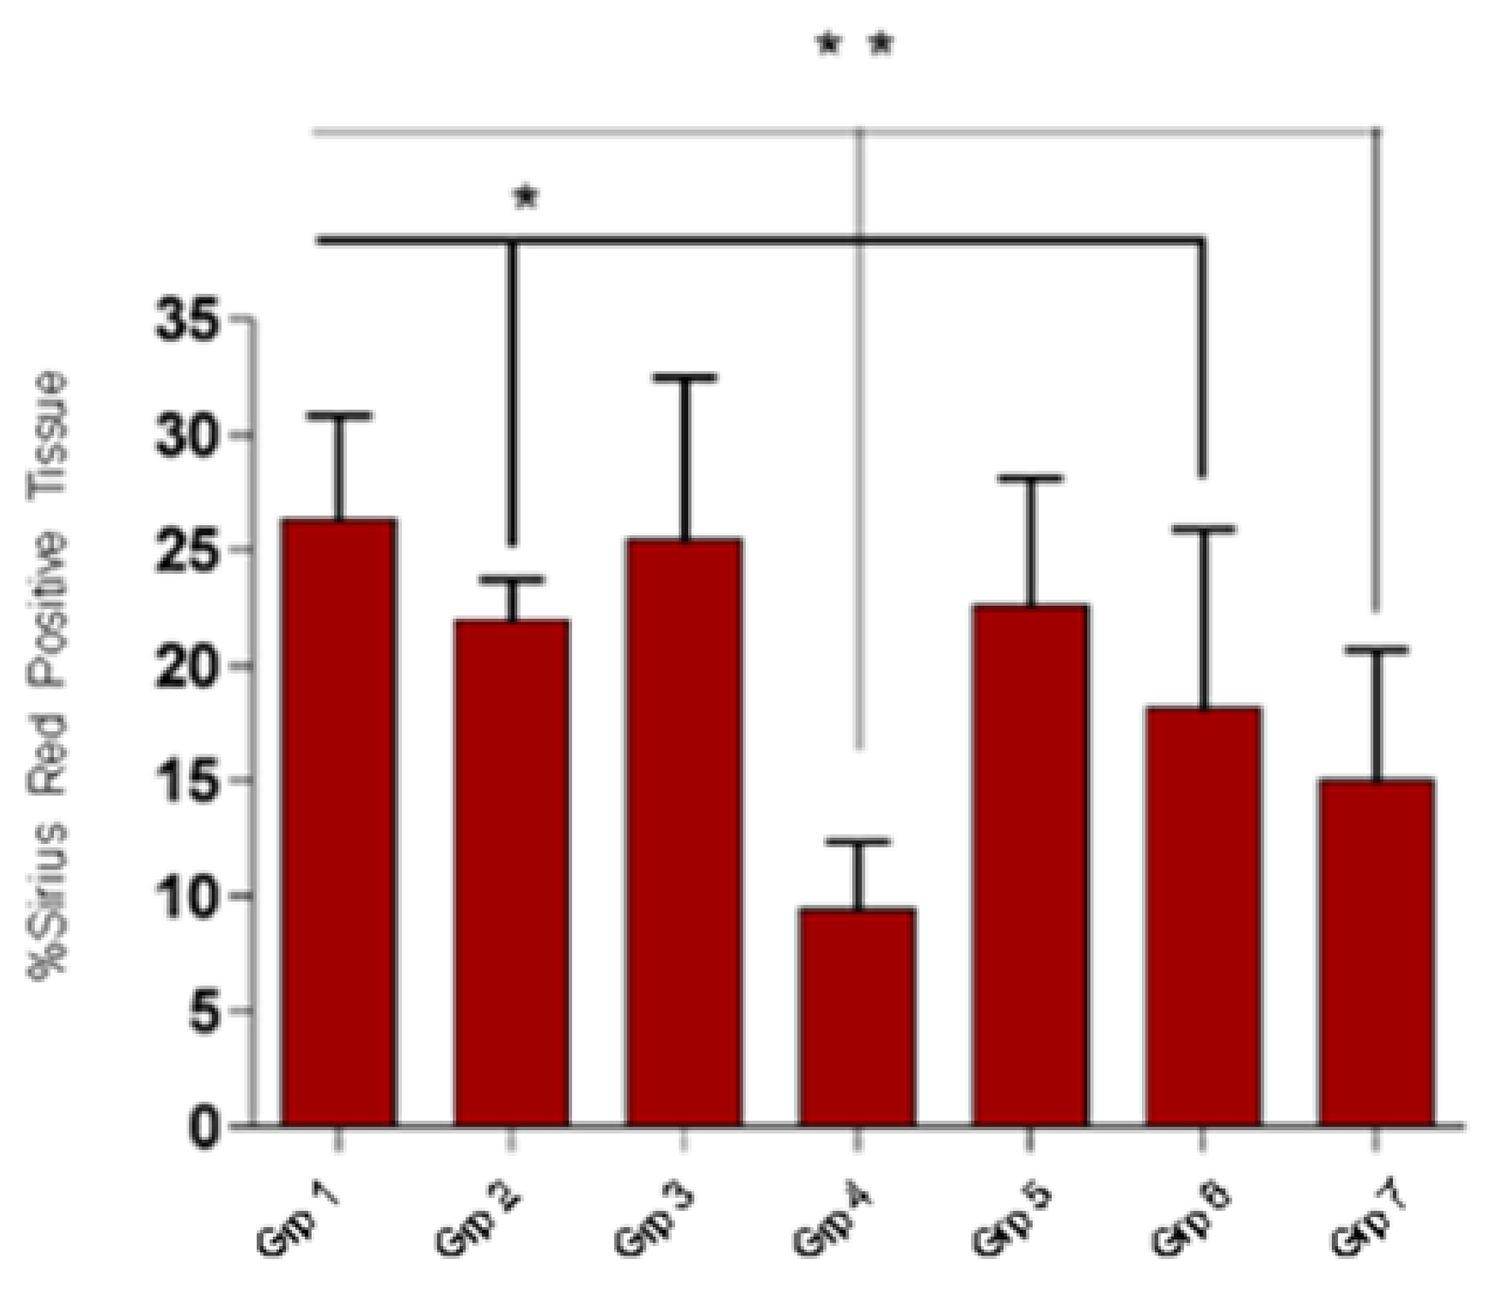
* *
*
%Sirius Red Positive Tissue
Grp 1
Grp 2
Grp 3
Grp 4
Grp 5
Grp 6
Grp 7

*P<0.05

**P<0.001

Histologic fibrosis scoring of Groups 1, 4 and 7*

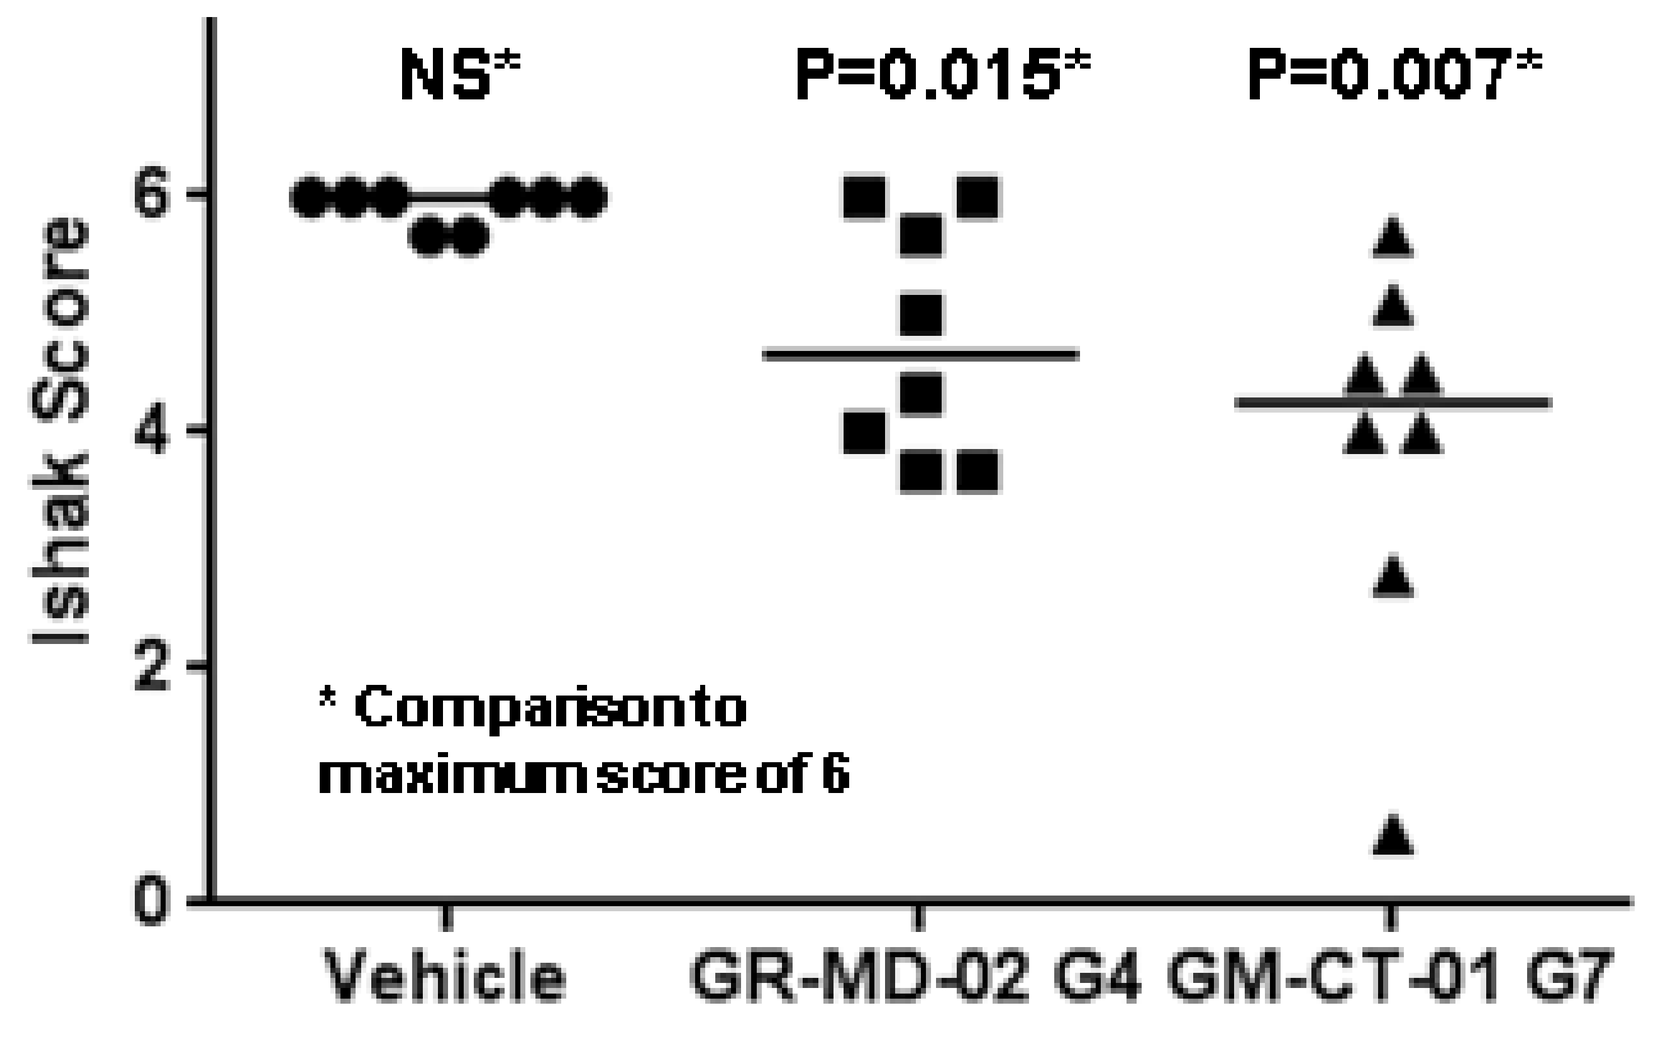
NS*
P=0.015*
P=0.007*
Ishak Score
* Comparison to maximum score of 6
Vehicle
GR-MD-02 G4
GM-CT-01 G7

* Slides were coded and then blindly evaluated by an expert liver pathologist. Each value represents the mean of 6 individual scores from each liver section.

Portal pressure measurements at sacrifice of TAA-treated rats for all treatment groups: GR-MD-02 reduced portal hypertension

Treatments	Portal pressure (cm H2O)
Control 0.9% NaCl (2x/week)	20.3±2.4
GR-MD-02 (60mg/kg 2x)	15.7*±2.9
GR-MD-02 (60mg/kg 1x)	18.9±1.4
GR-MD-02 (90mg/kg 1x)	17.1*±2.4
GM-CT-01 (105 mg/kg 2x)	20.8±1.9
GM-CT-01 (105 mg/kg 1x)	19.6±2.4
GM-CT-01 (180 mg/kg 1x)	18.5±3.7
Negative control	10.5**±2.4

P values compared to 'Control 0.9% NaCl'.
*p<0.05
**p<0.001

Comparison of biochemical measurements in TAA-treated rats

Treatment	% Collagen	COLI mRNA	COLI WB	α-SMA WB	AST	ALT
Control 0.9% NaCl (2x/week)	26±4.5	1	1±0.3	1±0.25	121.3±27.4	107.1±24.5
GR-MD-02 (60mg/kg 2x)	22*±1.8	1±0.37	0.45±0.05	0.786±0.41	112±58.3	96.9±15.8
GR-MD-02 (60mg/kg 1x)	25.5±7	0.69*±0.12	0.68±0.22	0.986±0.14	86*±20	88.1±15.5
GR-MD-02 (90mg/kg 1x)	9.5**±2.9	0.77*±0.18	0.288*±0.06	0.67±0.17	115±30.1	92.8±17.3
GM-CT-01 (105 mg/kg 2x)	23±5.5	0.93±0.33	0.83±0.4	1.2±0.28	108±19.1	92.7±16.6
GM-CT-01 (105 mg/kg 1x)	18*±7.7	1.1±0.26	0.35*±0.13	0.65±0.1	90.8*±11.2	82.3*±12.1
GM-CT-01 (180 mg/kg 1x)	15**±5.6	0.77±0.26	0.233*±0.17	0.47*±0.24	82.1*±15.13	76.8*±10.3
Negative control			0.06±0.01	0.14±0.01	79.8*±10.6	70.1*±7.7

P values compared to 'Control 0.9% NaCl'.
*p<0.05
**p<0.001

Summary and Conclusions:
- Treatment with two galectin protein inhibitors with different chemical compositions, but having common structural elements, significantly reduced fibrosis and reversed cirrhosis in a toxic model of liver fibrosis.
- Treatment effects on fibrosis and cirrhosis were associated with a reduction in portal hypertension.
- In vitro cell culture studies (data not shown) did not indicate a primary effect on LX-2 cells, a model of activated stellate cells, although GM-CT-01 did reduce alpha smooth muscle actin protein in livers.
- The reduction in collagen, reversal of cirrhosis, thinning of fibrous septa, and the presence of incomplete septa following once weekly administration of four doses suggests that the mechanism of galectin inhibition in this model may be activation of fibrosis resolution pathways.